KWS-2 – the Extended Q-Range / High-Flux SANS Diffractometer for Life Sciences

Aurel Radulescu, Jia-Jhen Kang, Marie-Sousai Appavou

Jülich Centre for Neutron Science (JCNS), Forschungszentrum Jülich GmbH, Garching, Germany

Instrument Performance

High Flux

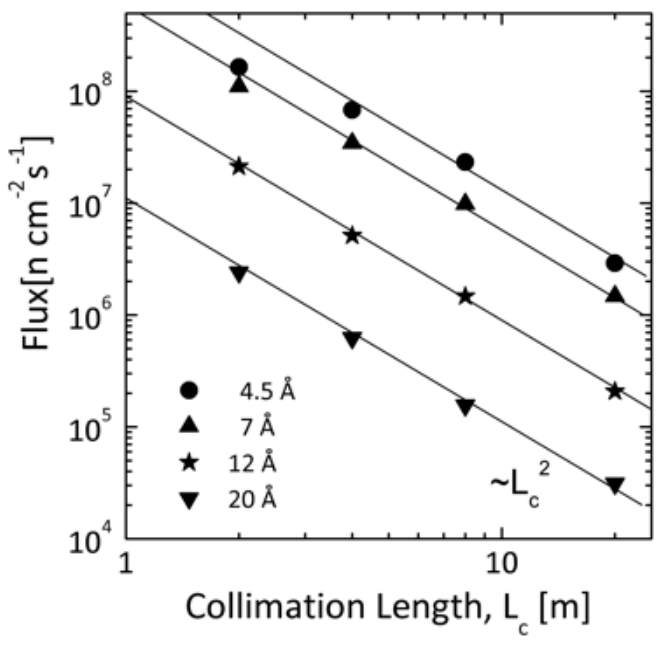

Flux[n cm^-2 s^-1]
4.5 Å
7 Å
12 Å
20 Å
~L_c^2
Collimation Length, L_c [m]

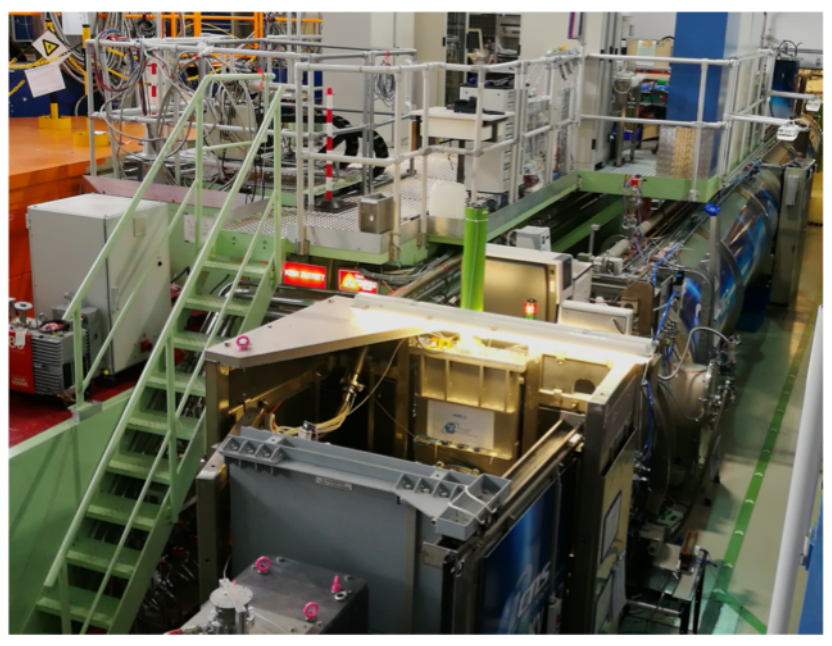

A. Radulescu et al. 2012 Nucl. Instr. Meth.
A. Radulescu et al. 2015 J. Appl. Cryst.
A. Radulescu et al. 2016 J. Vis. Exp
J. Houston et al. 2018 J. Appl. Cryst.
L. Balacescu et al. J. Appl. Cryst. (accepted)

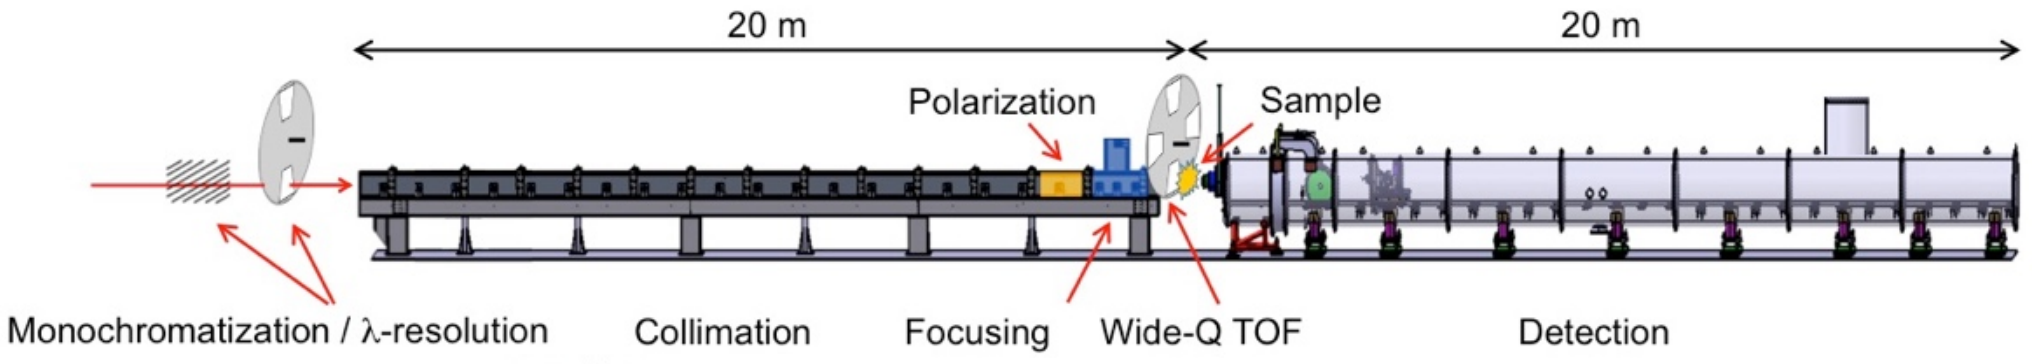

20 m
20 m
Polarization
Sample
Monochromatization / λ-resolution
Collimation
Focusing
Wide-Q TOF
Detection

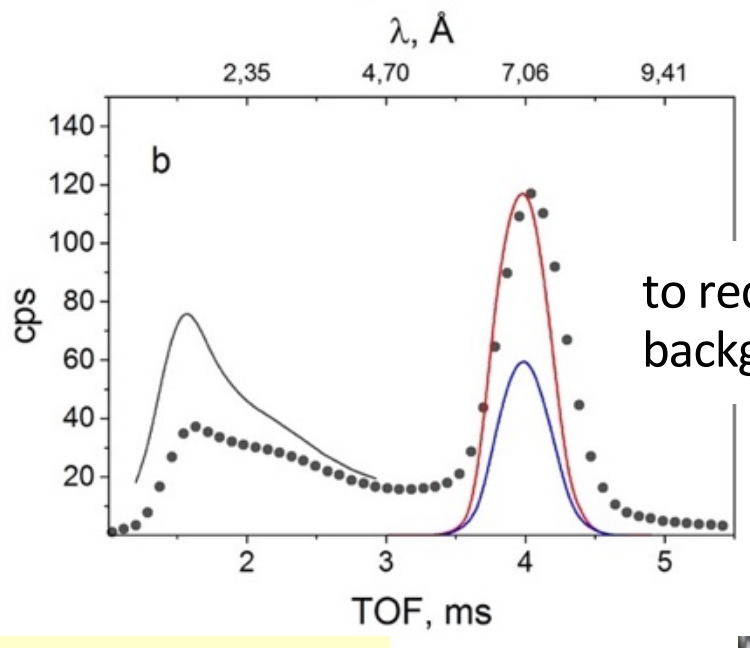

λ, Å
2,35
4,70
7,06
9,41
b
cps
TOF, ms

TOF – separation elastic / inelastic from hydrogenated samples

to reduce the incoherent background

Extended Q-range: $1x10^{-4}$ - $1Å^{-1}$

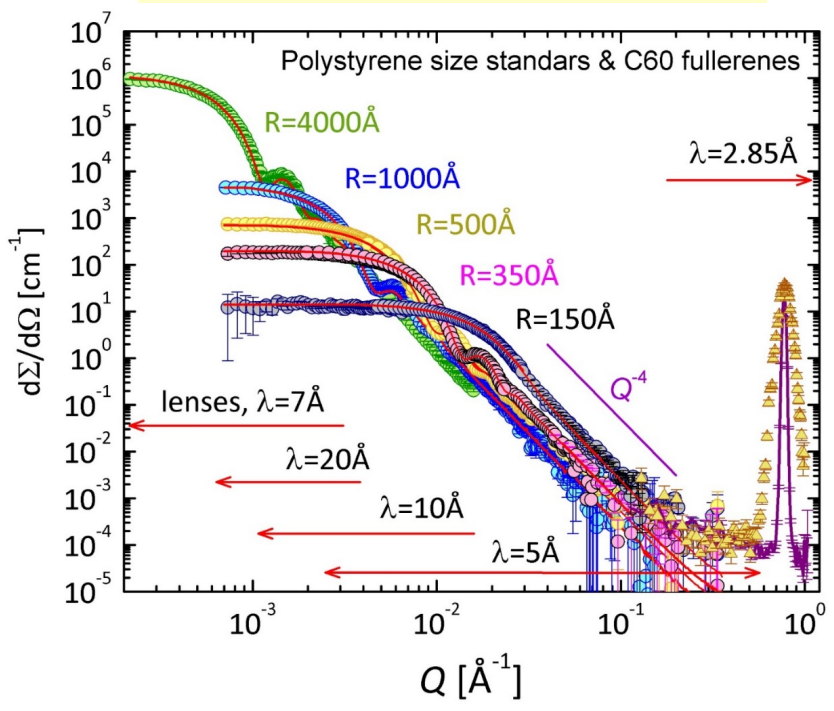

Polystyrene size standars & C60 fullerenes
R=4000Å
R=1000Å
R=500Å
R=350Å
R=150Å
λ=2.85Å
Q^-4
lenses, λ=7Å
λ=20Å
λ=10Å
λ=5Å
dΣ/dΩ [cm^-1]
Q [Å^-1]

Tunable resolution $\Delta\lambda/\lambda$: 2% - 20%

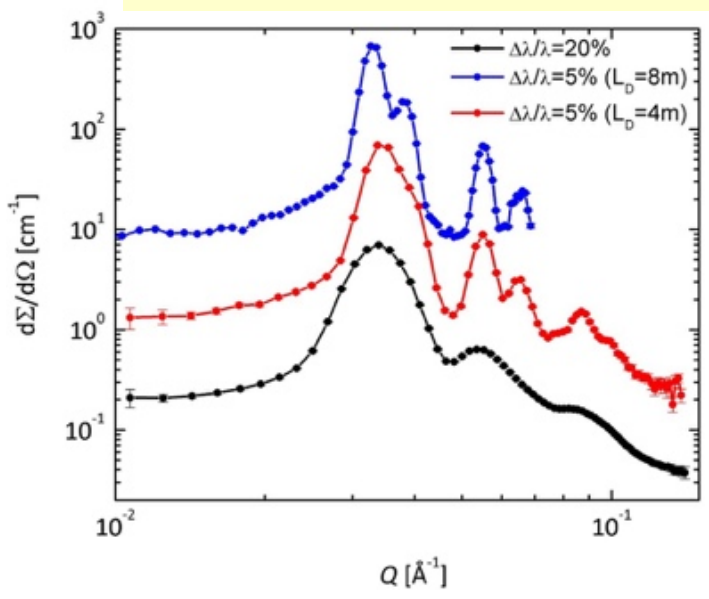

Δλ/λ=20%
Δλ/λ=5% (L_D=8m)
Δλ/λ=5% (L_D=4m)
dΣ/dΩ [cm^-1]
Q [Å^-1]

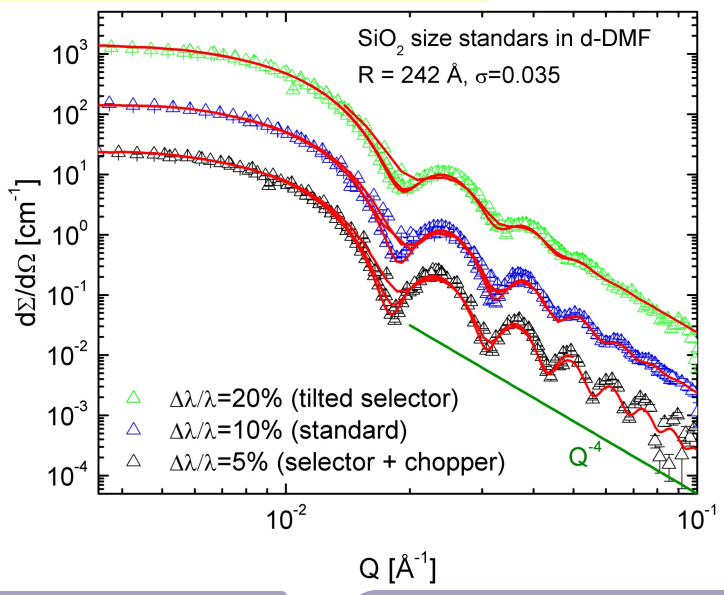

SiO2 size standars in d-DMF
R = 242 Å, σ=0.035
Δλ/λ=20% (tilted selector)
Δλ/λ=10% (standard)
Δλ/λ=5% (selector + chopper)
Q^-4
dΣ/dΩ [cm^-1]
Q [Å^-1]

Sample environments

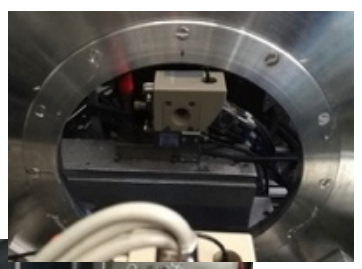

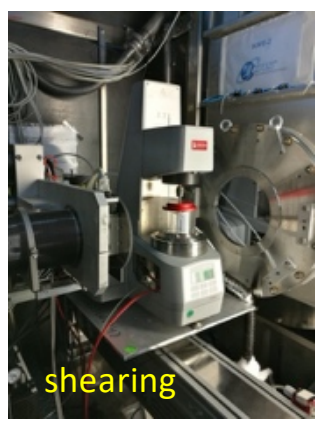

shearing

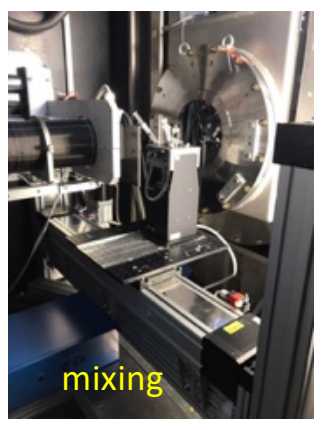

mixing

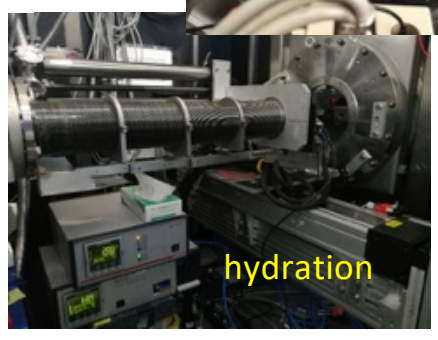

hydration

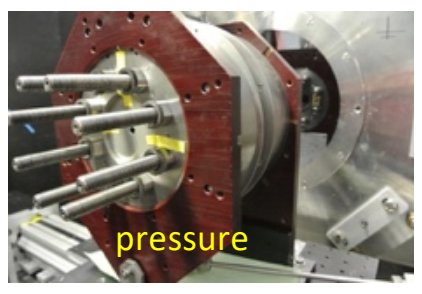

pressure

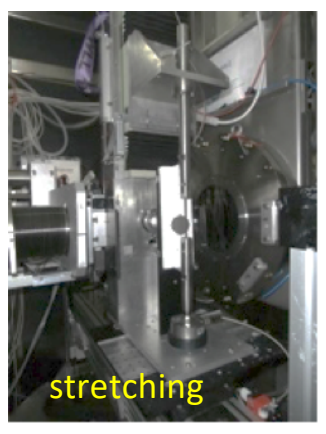

stretching

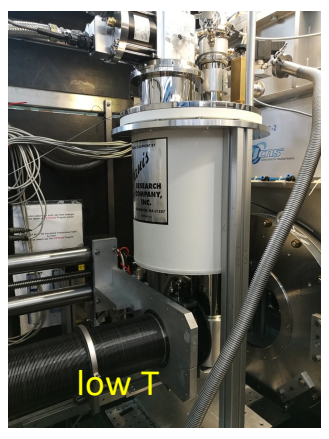

low T

In-situ complementarities – FTIR & DLS

in-situ FTIR

L. Balacescu et al., Appl. Sci. 2021

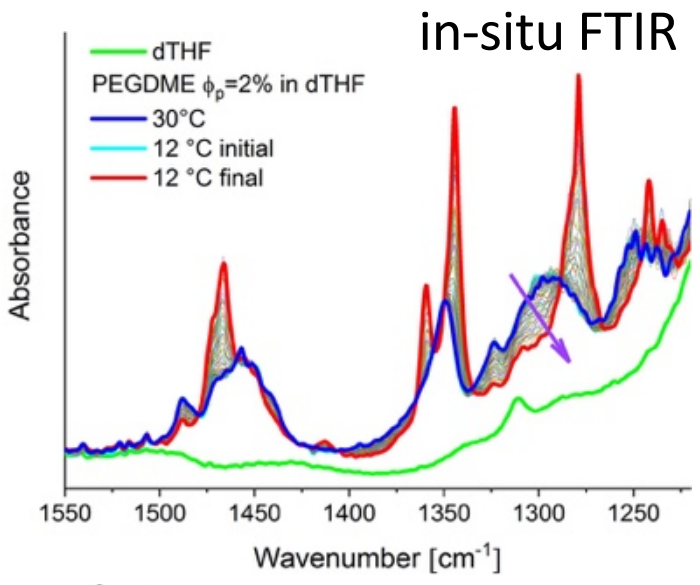

dTHF
PEGDME φ_p=2% in dTHF
30°C
12 °C initial
12 °C final
Absorbance
Wavenumber [cm^-1]

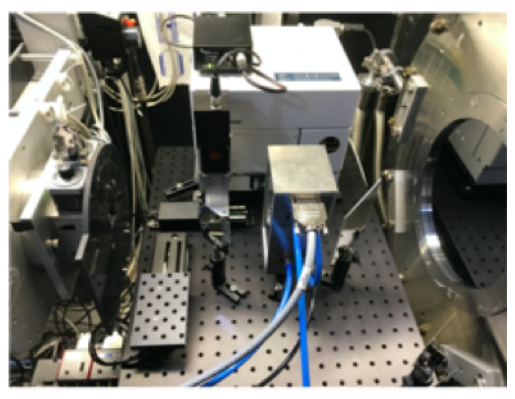

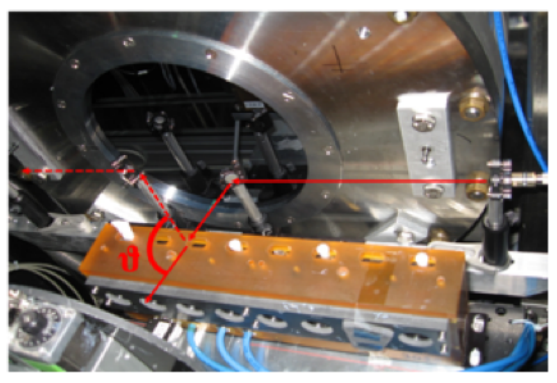

in-situ DLS

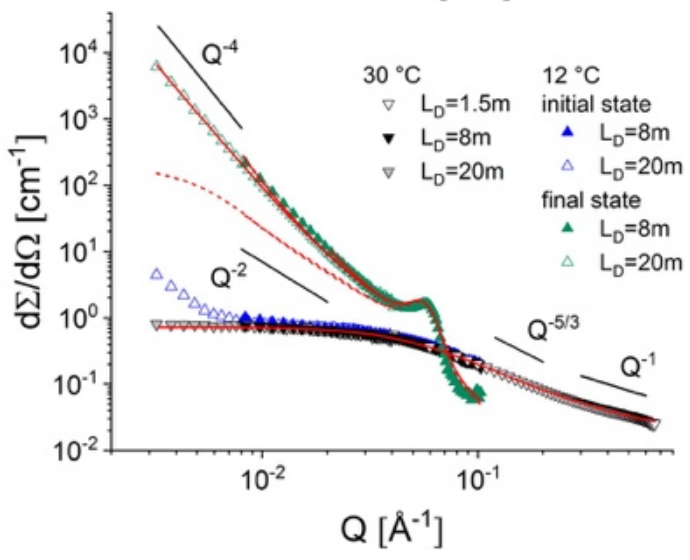

Q^-4
Q^-2
Q^-5/3
Q^-1
30 °C
L_D=1.5m
L_D=8m
L_D=20m
12 °C
initial state
L_D=8m
L_D=20m
final state
L_D=8m
L_D=20m
dΣ/dΩ [cm^-1]
Q [Å^-1]

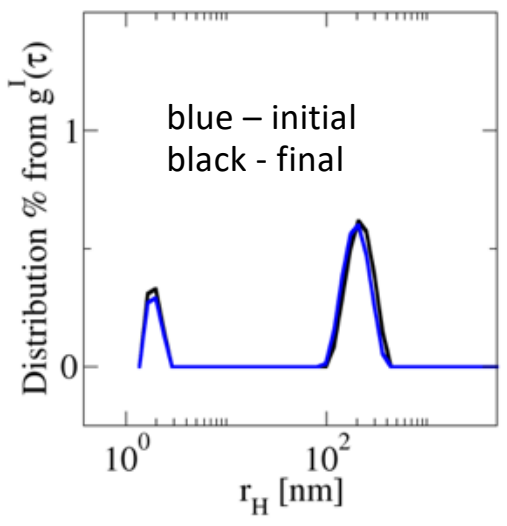

blue – initial
black - final
Distribution % from g^1(τ)
r_H [nm]

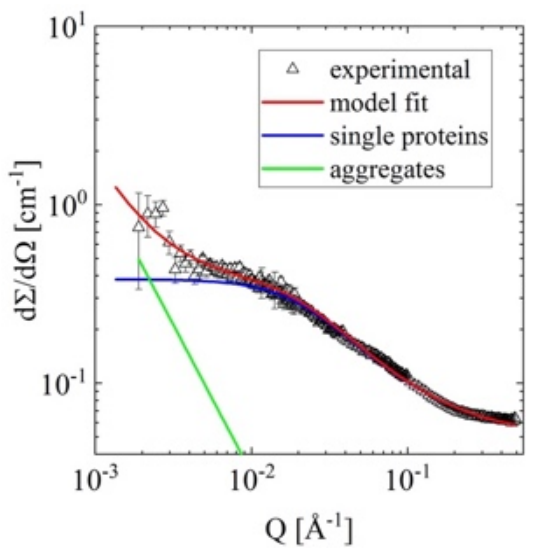

experimental
model fit
single proteins
aggregates
dΣ/dΩ [cm^-1]
Q [Å^-1]

- Monitoring the sample condition during SANS
- Observations at the local molecular level structure / conformation or large aggregates

Planned Upgrades

Robotics – continuous supply of instrument with samples

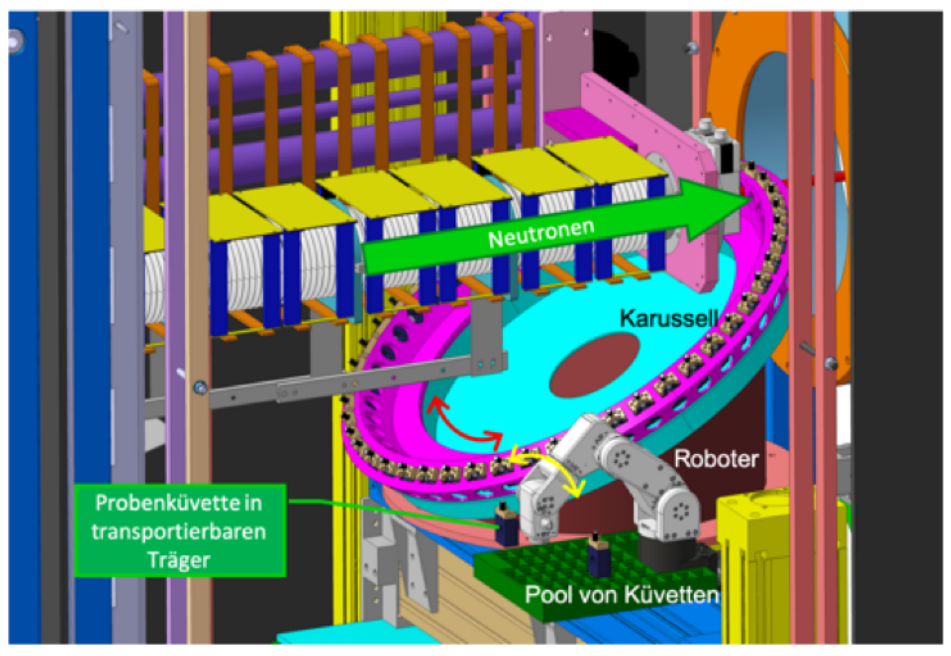

Neutronen
Karussell
Roboter
Probenküvette in transportierbaren Träger
Pool von Küvetten

Meca500 Robot (Mecademic Robotics) + thermostated Turret 48 positions (Quantum Northwest) + pool of cuvettes

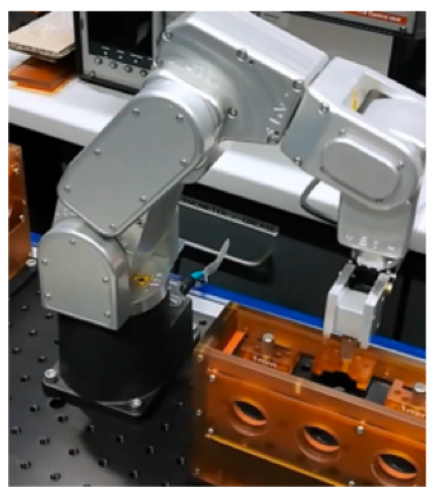

like TAIKAN, J-PARC; M. Hirai et al. J.Phys.Chem B 2019

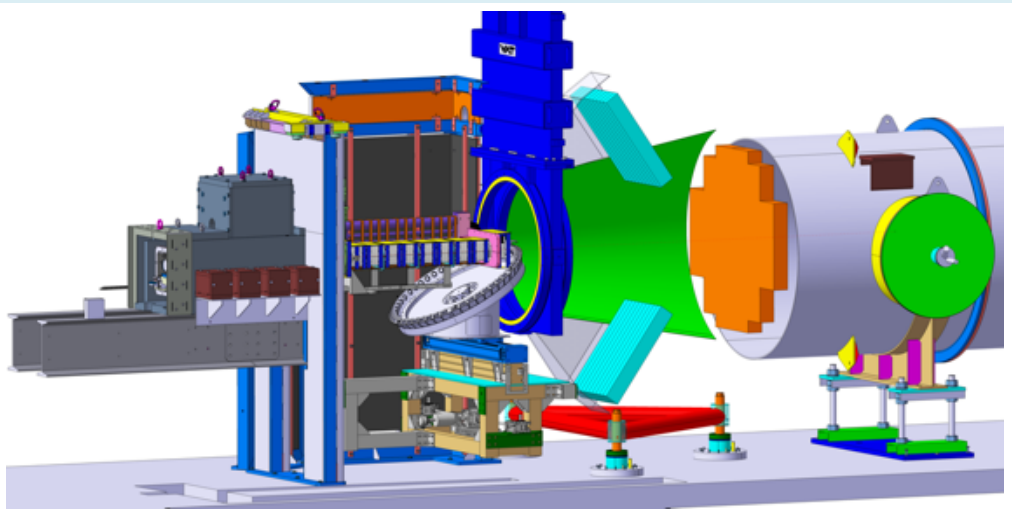

WANS – up to θ_s=50° (Q_{max} = 2 $Å^{-1}$)

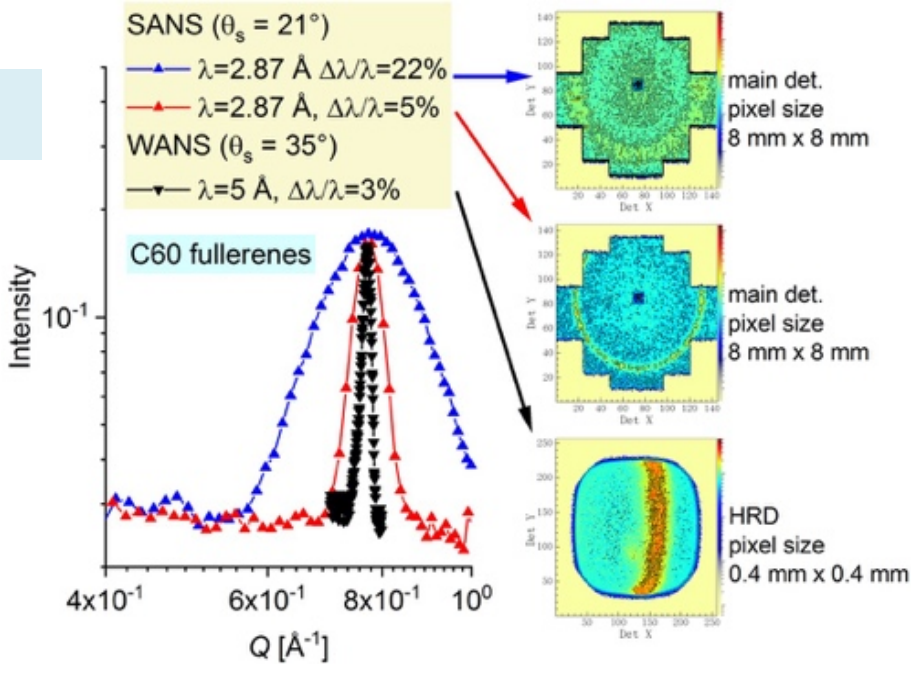

SANS (θ_s = 21°)
λ=2.87 Å Δλ/λ=22%
λ=2.87 Å, Δλ/λ=5%
WANS (θ_s = 35°)
λ=5 Å, Δλ/λ=3%
C60 fullerenes
Intensity
Q [Å^-1]
main det.
pixel size
8 mm x 8 mm
main det.
pixel size
8 mm x 8 mm
HRD
pixel size
0.4 mm x 0.4 mm

Examples

Extended Q-range – multiple structural levels: morphologies

B. Krugmann et al., Sci. Rep. 2020

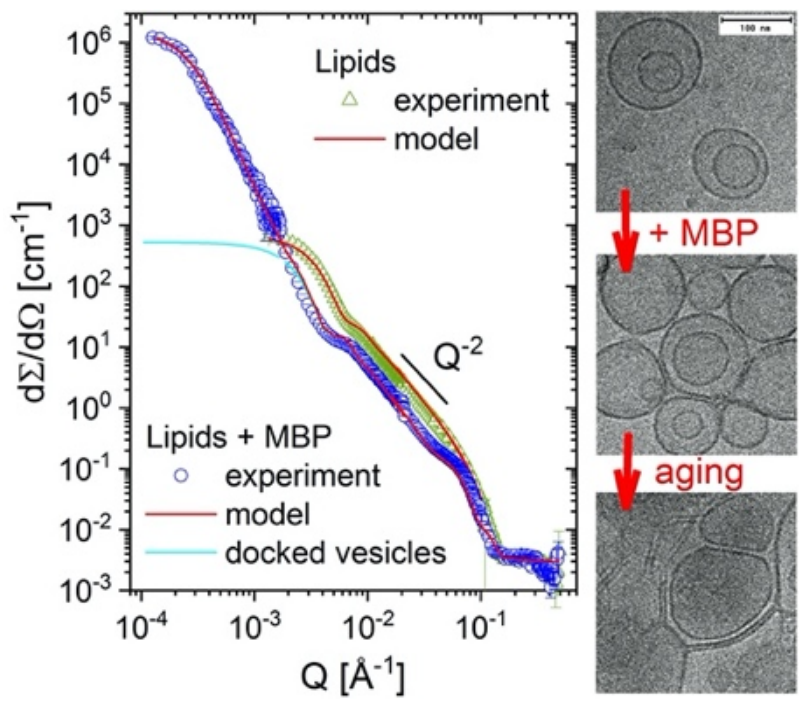

Lipids
experiment
model
Q^-2
Lipids + MBP
experiment
model
docked vesicles
dΣ/dΩ [cm^-1]
Q [Å^-1]
+ MBP
aging

Contrast variation – drug carriers (LNPs)

N. Davies et al., Molecular Therapy Nucleic Acids, 2021

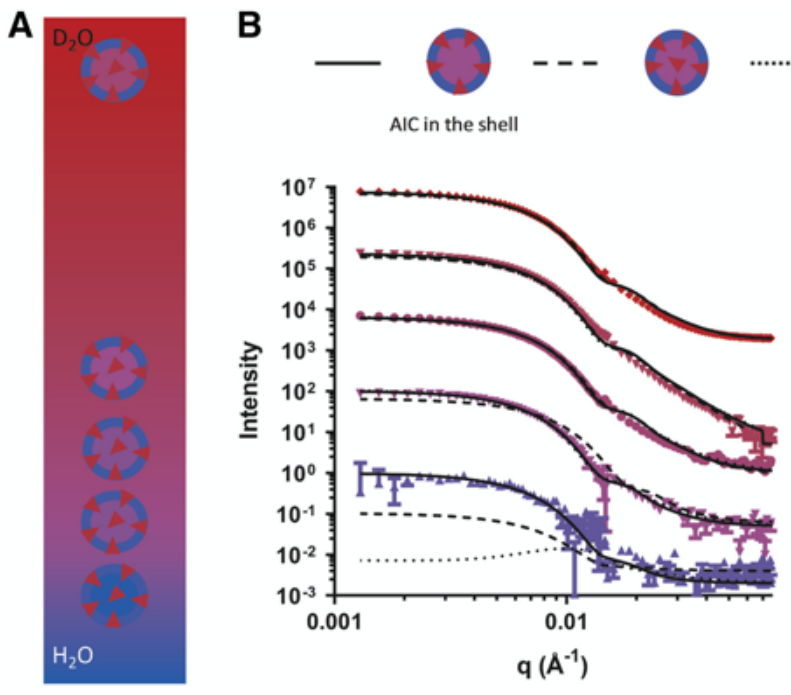

A
B
D2O
H2O
AIC in the shell
AIC in the core
100% D2O
50% D2O
35% D2O
25% D2O
15% D2O
Intensity
q (Å^-1)

High Intensity – low concentration protein solutions

J. Houston et al., J. Appl. Cryst. 2018

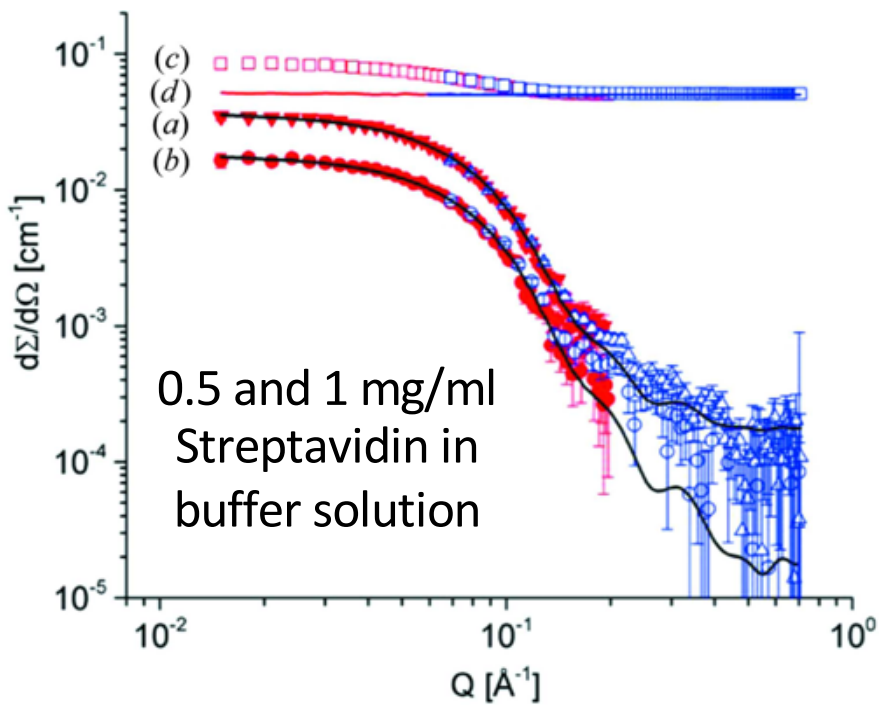

(c)
(d)
(a)
(b)
0.5 and 1 mg/ml Streptavidin in buffer solution
dΣ/dΩ [cm^-1]
Q [Å^-1]

SEC – SANS

flow-through cell + holder: C. Dicko et al., Rev. Sci. Inst. 2020

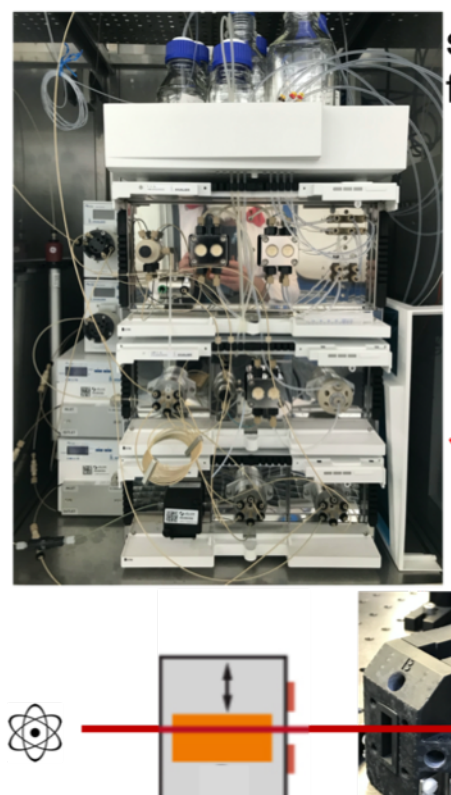

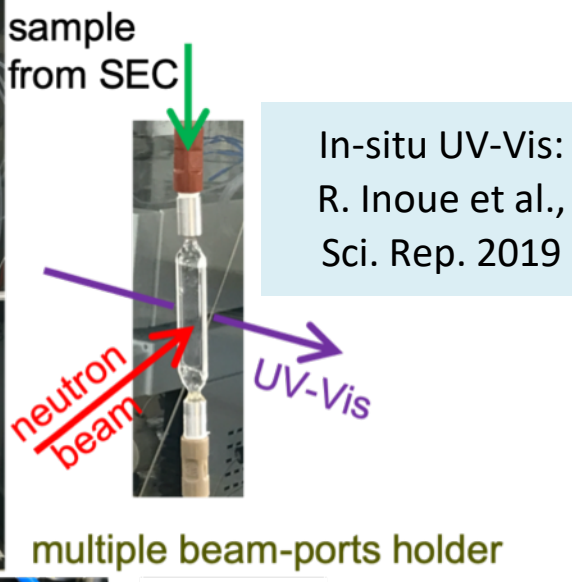

sample from SEC
neutron beam
UV-Vis

In-situ UV-Vis: R. Inoue et al., Sci. Rep. 2019

multiple beam-ports holder

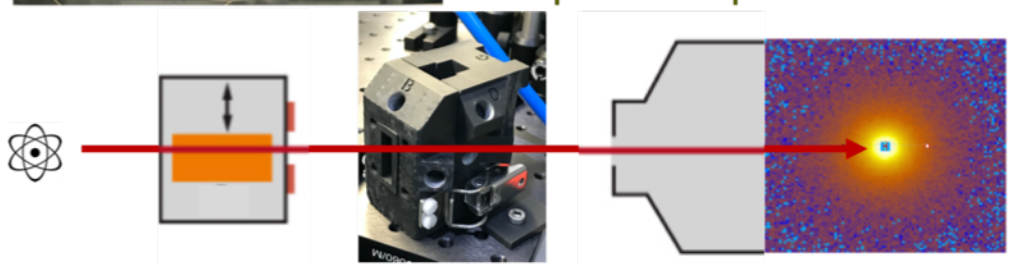

Contact: A. Radulescu
E-mail: a.radulescu@fz-juelich.de
Phone: +49.89.289.10712